SAFETY AND EFFICACY OF BEMPEDOIC ACID IN PATIENTS WITH RENAL IMPAIRMENT

Peter P. Toth[1], Alberico L. Catapano[2], Lei Lei[3], Michael J. Louie[3], George Bakris[4]

[1]CGH Medical Center, Sterling, IL, and The Ciccarone Center for the Prevention of Cardiovascular Disease, Johns Hopkins University School of Medicine, Baltimore, MD; [2]Department of Pharmacological and Biomolecular Sciences, University of Milan, Milan, Italy and IRCCS MultiMedica, Milan, Italy; [3]Esperion Therapeutics, Inc., Ann Arbor, MI; [4]University of Chicago Medicine, Chicago, IL

Bempedoic acid significantly lowered low-density lipoprotein cholesterol and was well tolerated in patients with renal impairment

Background

- Renal impairment and elevated levels of low-density lipoprotein cholesterol (LDL-C) are both associated with cardiovascular risk[1]
- Patients with chronic kidney disease (CKD) are also at high risk of medication adverse events, possibly due to reduced renal excretion, as well as the likelihood of polypharmacy and high propensity for comorbidities[2]
- Lowering LDL-C reduces the risk of cardiovascular events in patients with CKD.[3] Statins are utilized to lower LDL-C in this population[2]; however, patients may not reach lipid goals without additional lipid-lowering therapies (LLT)
- Bempedoic acid is an oral ATP-citrate lyase inhibitor that significantly lowers LDL-C levels in patients with hypercholesteremia[4–7] and can be used without dose adjustment for patients with mild or moderate renal impairment[8]

Objective

- To investigate the safety and efficacy of bempedoic acid in patients with impaired renal function by a pooled analysis of data from four phase 3 trials

Methods

Study design and treatment

- Pooled data from four phase 3 studies (CLEAR Harmony, CLEAR Wisdom, CLEAR Serenity, and CLEAR Tranquility [NCT02666664, NCT02991118, NCT02988115, and NCT03001076]); patients were randomized 2:1 to receive either bempedoic acid 180 mg or placebo once daily plus stable background LLT[4–7]

Patients

- Patients were grouped according to their respective renal function, as determined by baseline estimated glomerular filtration rate (eGFR [mL/min/1.73 m²]): stage 1 (≥90); stage 2 (60 to <90); and stage 3a+b (30 to <60). Calculation of eGFR was performed using the Modification of Diet in Renal Disease (MDRD) formula

Assessments

- Percent change in LDL-C at week 12 was the primary efficacy endpoint
 - LDL-C values were calculated using either the Friedewald formula or measured directly using the Multigent Direct LDL assay (Architect system, Abbott; when triglycerides >400 mg/dL or LDL-C <50 mg/dL)
- Percent changes in total cholesterol (TC), non–high-density lipoprotein cholesterol (non–HDL-C), apolipoprotein B (Apo B), and high-sensitivity C-reactive protein (hsCRP) were secondary efficacy endpoints
- Quantification of lipids and biomarkers was performed at a central laboratory (Q2 Solutions)
- Safety assessments were investigator reported and included treatment-emergent adverse events (TEAEs), serious TEAEs, and clinical laboratory results

Statistical analysis

- The efficacy analysis population included all patients who were randomized to treatment; the safety analysis population included all patients receiving at least one dose
- Least squares (LS) means, 95% confidence intervals (CIs), and *P* values are based on an analysis of covariance (ANCOVA) with percent change from baseline as the dependent variable, study and treatment as fixed factors, and baseline as a covariate. Comparisons of hsCRP are based on a nonparametric method
- The interaction *P* value was obtained from the interaction term via an ANCOVA with percent change from baseline as the dependent variable (log-transformed for hsCRP), study, treatment, eGFR category, and interaction term between treatment and eGFR category as fixed factors, and baseline as a covariate

Results

Participants

- Only patients with impaired renal function are presented
- 2833 patients were included in the efficacy analysis; 81% had stage 2 renal function, and 19% had stage 3a+b
- Within each renal function stage, baseline measurements of TC, LDL-C, non–HDL-C, Apo B, and hsCRP, as well as demographics, were similar between the treatment and placebo groups (**Table 1**)

Table 1. Patient Demographics and Baseline Characteristics*

Renal function stage	Stage 2		Stage 3a+b	
	Bempedoic acid n = 1533	Placebo n = 758	Bempedoic acid n = 359	Placebo n = 183
Age, years	65.4 (8.8)	66.5 (8.3)	70.7 (7.7)	70.0 (7.5)
Male, n (%)	1019 (66.5)	494 (65.2)	205 (57.1)	98 (53.6)
Race, n (%)				
White	1460 (95.2)	716 (94.5)	352 (98.1)	177 (96.7)
Black or African American	47 (3.1)	30 (4.0)	5 (1.4)	5 (2.7)
Asian	18 (1.2)	7 (0.9)	2 (0.6)	0
Other	8 (0.5)	5 (0.7)	0	1 (0.6)
Non-Hispanic, n (%)	1468 (95.8)	725 (95.7)	345 (96.1)	175 (95.6)
BMI, kg/m²	29.7 (5.0)	30.0 (5.1)	30.2 (5.2)	30.0 (5.0)
TC, mg/dL	193.0 (42.6)	192.2 (42.9)	194.5 (44.7)	194.2 (45.4)
LDL-C, mg/dL	114.0 (35.4)	112.8 (35.2)	113.2 (37.5)	113.7 (39.0)
Non–HDL-C, mg/dL	142.6 (40.8)	141.8 (40.8)	145.2 (42.9)	144.8 (43.6)
Apo B, mg/dL	101.5 (30.8)	100.7 (31.0)	101.6 (31.0)	102.8 (32.5)
hsCRP, mg/L**	1.6 (0.8, 3.5)	1.6 (0.9, 3.5)	2.1 (1.1, 4.3)	2.4 (1.1, 5.5)

Apo B = apolipoprotein B; BMI = body mass index; hsCRP = high-sensitivity C-reactive protein; LDL-C = low-density lipoprotein cholesterol; non–HDL-C = non–high-density lipoprotein cholesterol; TC = total cholesterol.
Data presented are mean (standard deviation) unless otherwise indicated.
*3623 patients were randomized; 2 patients did not receive a dose, so 3621 patients were in the safety analysis. Four patients with stage 4 renal function and 786 patients with stage 1 renal function were excluded from the presentation. **Median (quartile 1, quartile 3). Stages defined by estimated glomerular filtration rate (mL/min/1.73 m²): 60 to <90 (stage 2) and 30 to <60 (stage 3a+b).

Primary endpoint: percent change in LDL-C

- Significant LDL-C lowering was associated with bempedoic acid treatment, and mean reductions were significantly greater than placebo (*P* <0.0001 for both renal function stages) and similar across renal function stages (interaction *P* = 0.4442) (**Figure 1**)

Figure 1. LDL-C Mean Percent Change at Week 12 by Treatment Group and Renal Function Stage

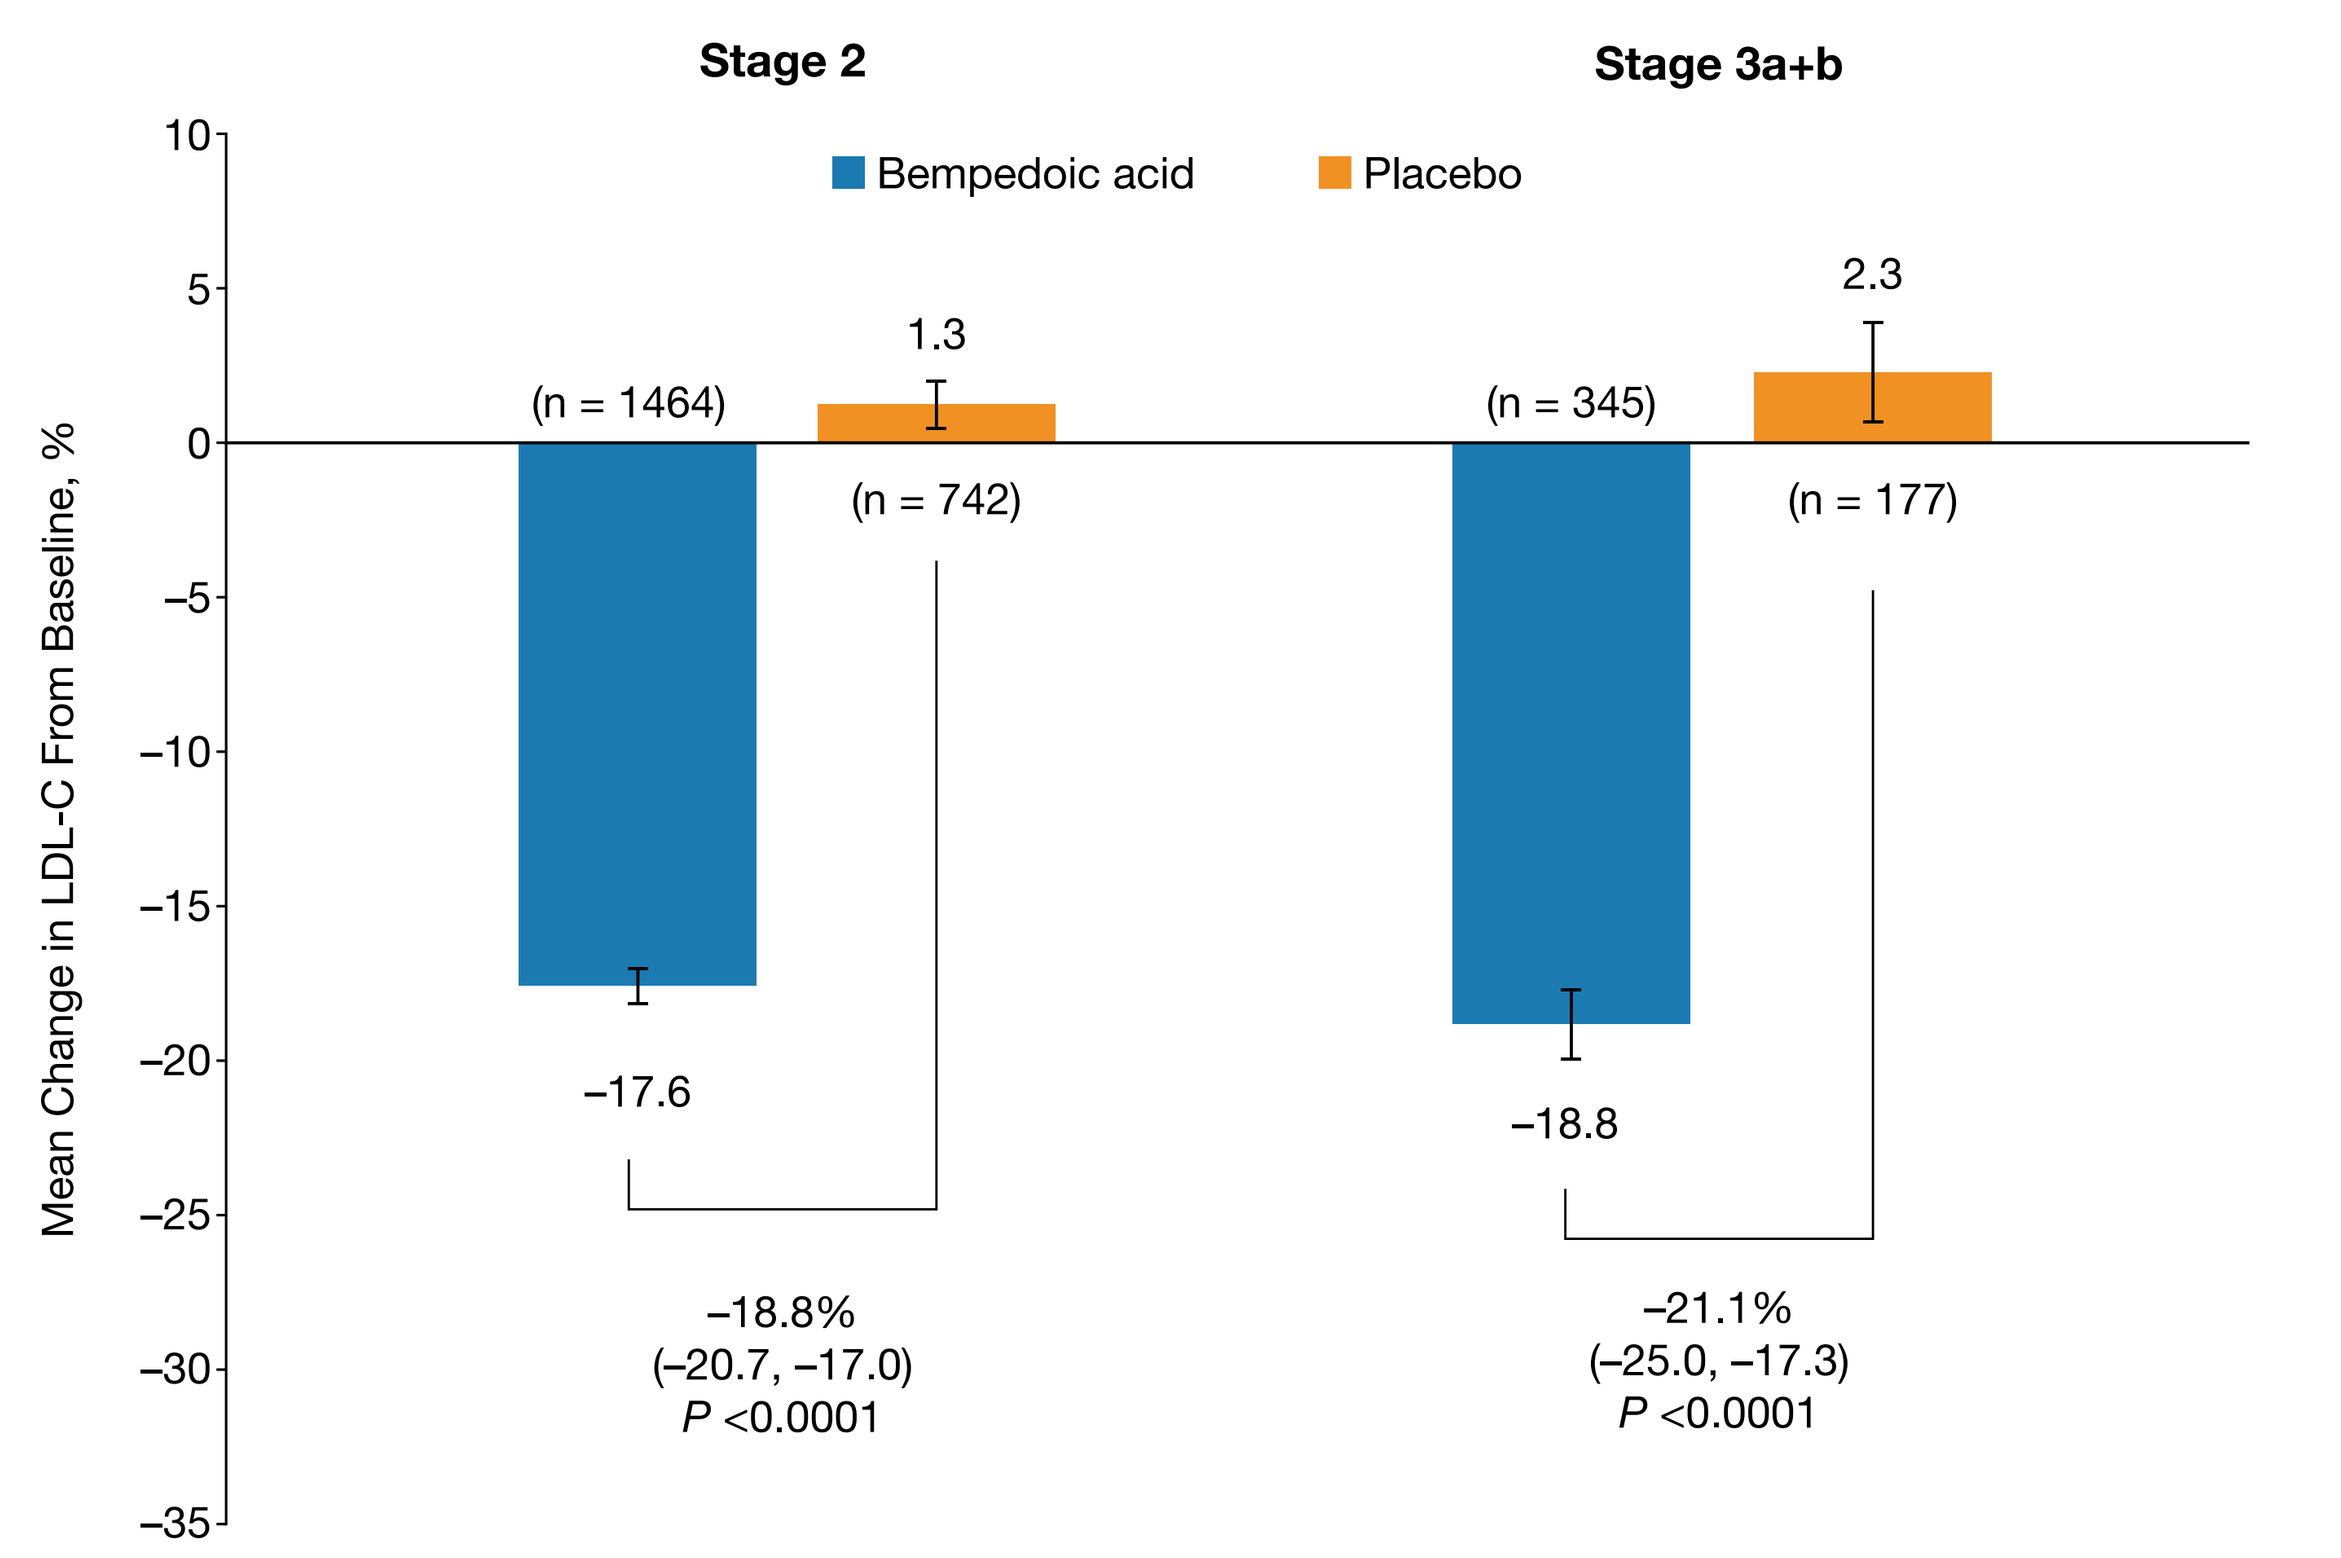
Stage 2
Stage 3a+b
Bempedoic acid
Placebo
Mean Change in LDL-C From Baseline, %
10
5
0
−5
−10
−15
−20
−25
−30
−35
(n = 1464)
1.3
(n = 742)
−17.6
−18.8%
(−20.7, −17.0)
P <0.0001
(n = 345)
2.3
(n = 177)
−18.8
−21.1%
(−25.0, −17.3)
P <0.0001

LDL-C = low-density lipoprotein cholesterol.
Data presented are least squares means, their difference (95% confidence interval).
Stages defined by estimated glomerular filtration rate (mL/min/1.73 m²): 60 to <90 (stage 2) and 30 to <60 (stage 3a+b).

Secondary endpoints: percent change in TC, non–HDL-C, Apo B, and hsCRP

- The placebo-corrected LS mean (95% CI) percent change in TC, non–HDL-C, and Apo B were statistically significant irrespective of renal impairment (interaction *P* = 0.7371, 0.6843, and 0.8706, respectively) at week 12 (**Table 2**)
- The location shift for hsCRP was statistically significant irrespective of renal impairment (interaction *P* = 0.2173 in the log scale) at week 12 (**Table 2**)

Table 2. Percent Changes From Baseline to Week 12 in TC, Non–HDL-C, Apo B, and hsCRP With Bempedoic Acid by Renal Function Stage

	Stage 2		Stage 3a+b	
	Bempedoic acid	Placebo	Bempedoic acid	Placebo
TC, n	1468	742	346	177
LS mean (SE)	−11.6 (0.4)	0.6 (0.5)	−10.6 (0.9)	1.2 (1.2)
Placebo-corrected LS mean (95% CI)	−12.2 (−13.5, −10.9)*		−11.8 (−14.6, −9.0)*	
Non–HDL-C, n	1466	742	346	177
LS mean (SE)	−13.3 (0.5)	1.4 (0.7)	−11.9 (1.1)	2.2 (1.5)
Placebo-corrected LS mean (95% CI)	−14.7 (−16.3, −13.0)*		−14.0 (−17.8, −10.3)*	
Apo B, n	1443	729	340	172
LS mean (SE)	−10.7 (0.4)	3.0 (0.7)	−8.8 (1.0)	3.8 (1.4)
Placebo-corrected LS mean (95% CI)	−13.7 (−15.3, −12.1)*		−12.5 (−16.0, −9.0)*	
hsCRP, n	1440	731	338	173
Median (Q1, Q3)	−23.0 (−51.0, 21.5)	−4.1 (−34.0, 49.1)	−29.4 (−57.8, 7.3)	−4.2 (−34.8, 58.3)
Location shift (95% CI)	−18.0 (−23.0, −13.0)*		−25.6 (−35.7, −15.3)*	

Apo B = apolipoprotein B; CI = confidence interval; hsCRP = high-sensitivity C-reactive protein; LS = least squares; non–HDL-C = non–high-density lipoprotein cholesterol; Q = quartile; SE = standard error; TC = total cholesterol. **P* <0.0001. Stages defined by estimated glomerular filtration rate (mL/min/1.73 m²): 60 to <90 (stage 2) and 30 to <60 (stage 3a+b).

Safety endpoints

- The overall pattern of adverse events was consistent between both renal function stages; any differences appeared to be driven by overall bempedoic acid or placebo differences rather than by renal function stage at baseline (**Table 3**)

Table 3. TEAEs by Baseline Renal Function Stage

n (%)	Stage 2		Stage 3a+b	
	Bempedoic acid n = 1532	Placebo n = 758	Bempedoic acid n = 359	Placebo n = 183
Any TEAE	1126 (73.5)	547 (72.2)	275 (76.6)	142 (77.6)
Any serious TEAE	201 (13.1)	97 (12.8)	72 (20.1)	28 (15.3)
Any TEAE related to IMP	374 (24.4)	164 (21.6)	100 (27.9)	36 (19.7)
Any TEAE leading to IMP discontinuation	171 (11.2)	56 (7.4)	47 (13.1)	19 (10.4)
Any TEAE with fatal outcome	12 (0.8)	2 (0.3)	3 (0.8)	1 (0.5)
TEAEs ≥5% in any group				
UTI	68 (4.4)	36 (4.7)	21 (5.8)	16 (8.7)
Anemia	27 (1.8)	10 (1.3)	18 (5.0)	5 (2.7)
Diarrhea	55 (3.6)	16 (2.1)	15 (4.2)	11 (6.0)
Arthralgia	71 (4.6)	32 (4.2)	11 (3.1)	10 (5.5)
Nasopharyngitis	126 (8.2)	64 (8.4)	15 (4.2)	9 (4.9)
Myalgia	77 (5.0)	39 (5.1)	14 (3.9)	8 (4.4)
Fatigue	38 (2.5)	23 (3.0)	5 (1.4)	10 (5.5)

IMP = investigational medicinal product; TEAE = treatment-emergent adverse event; UTI = urinary tract infection.
Stages defined by estimated glomerular filtration rate (mL/min/1.73 m²): 60 to <90 (stage 2) and 30 to <60 (stage 3a+b).

- Creatinine levels at week 52 were generally consistent within each renal function stage in both bempedoic acid and placebo arms (**Table 4**); over 52 weeks, mean creatinine in each arm varied ≤7% at each assessed time point

Table 4. Creatinine and Percent Change from Baseline at Week 52, by Treatment Group and Renal Function Stage

Mean (SD)	Stage 2		Stage 3a+b	
	Bempedoic acid n = 1158	Placebo n = 581	Bempedoic acid n = 271	Placebo n = 131
Creatinine, mg/dL	0.98 (0.18)	0.94 (0.16)	1.34 (0.34)	1.25 (0.35)
Percent change from baseline	1.96 (12.84)	−1.53 (11.22)	5.59 (17.81)	−0.56 (13.06)

SD = standard deviation.
Stages defined by estimated glomerular filtration rate (mL/min/1.73 m²): 60 to <90 (stage 2) and 30 to <60 (stage 3a+b).

- 0.3% of patients treated with bempedoic acid showed an increase >1 mg/dL creatinine from baseline at any time point during the studies (**Table 5**)

Table 5. Incidence of Changes in Renal Laboratory Values

n (%)	Bempedoic acid (n = 2424)			Placebo (n = 1197)		
	Patients with elevation	Patients with elevation and clinical symptoms	Patients with elevation and discontinuation of IMP due to any AE	Patients with elevation	Patients with elevation and clinical symptoms	Patients with elevation and discontinuation of IMP due to any AE
Increase from baseline in creatinine >1 mg/dL	7 (0.3)	2 (<0.1)	0	1 (<0.1)	0	0
Increase from baseline in creatinine >30% within 4 weeks of first dose of IMP	45 (1.9)	1 (<0.1)	4 (0.2)	5 (0.4)	0	0
eGFR <15 mL/min/1.73 m²	2 (<0.1)*	0	0	0	0	0

AE = adverse event; eGFR = estimated glomerular filtration rate; IMP = investigational medicinal product. *Both patients had baseline stage 3a+b renal function.

Conclusions

- Bempedoic acid was generally well tolerated in this large group of patients with eGFR between 30 and <90 mL/min/1.73 m²
 - The most common TEAEs (≥5% in any group) included urinary tract infection, anemia, diarrhea, arthralgia, nasopharyngitis, myalgia, and fatigue
- Creatinine changes with bempedoic acid were generally small across the pooled phase 3 data and reversible on discontinuation
- Bempedoic acid significantly lowered LDL-C regardless of renal function stage
- Patients with severe renal impairment (eGFR <30 mL/min/1.73 m²) or end-stage renal disease receiving dialysis were not recruited for these trials, which is a limitation of the study

References

1. Waters DD. *Curr Opin Lipidol*. 2015;26:195–199.
2. Kidney Disease: Improving Global Outcomes (KDIGO) Lipid Work Group. *Kidney Int Suppl*. 2013; 3:259–305.
3. Baigent C, et al. *Lancet*. 2011;377:2181–2192.
4. Ray KK, et al. *N Engl J Med*. 2019;380:1022–1032.
5. Laufs U, et al. *J Am Heart Assoc*. 2019;8:e011662.
6. Ballantyne CM, et al. *Atherosclerosis*. 2018;277:195–203.
7. Goldberg AC, et al. *JAMA*. 2019;322:1780–1788.
8. NEXLETOL® (bempedoic acid) [package insert]. Ann Arbor, MI: Esperion Therapeutics, Inc.; September 2021.

Acknowledgments and Disclosures

We thank all investigators and patients who participated in these trials. We thank Bethany Helms for her contribution to the study.

Clinical trials included in this analysis were funded by Esperion Therapeutics, Inc. Esperion participated in the study design, research, data collection, analysis, and interpretation of data, and writing, reviewing, and approving the poster for presentation. All authors had access to the data and participated in the development, review, and approval of the poster.

Medical writing and editorial assistance were provided by Laurel Riemann, PharmD, and Charlotte Watson of Spark Medica Inc., funded by Esperion Therapeutics, Inc.

PPT has received honoraria for consultancy from Amarin, AstraZeneca, Kowa, Merck, Novo Nordisk, and Resverlogix.

ALC has received funding/grant support and honoraria for consultancy from Aegerion, Amgen, Amryt, AstraZeneca, Bayer, Daiichi Sankyo, Eli Lilly, Genzyme, Kowa, Mediolanum, Menarini, Merck, Mylan, Novartis, Pfizer, Recordati, Regeneron, Sanofi, and Sigma-Tau.

BH was an employee of Esperion Therapeutics, Inc., during the conduct of this research; she is now an employee of Pfizer, Inc.

LL is a consultant for Esperion Therapeutics, Inc.

MJL is a current employee of Esperion Therapeutics, Inc., and may own Esperion stock and/or stock options.

GB has received honoraria for consultancy from Alnylam, AstraZeneca, Bayer, Horizon, Ionis, KBP Biosciences, Merck, Novo Nordisk, Quantum Genomics, and Vascular Dynamics.

Presented at the Cardiometabolic Health Congress, October 19–22, 2022.