EFFICACY AND SAFETY OF BEMPEDOIC ACID IN PATIENTS WITH METABOLIC SYNDROME

Michael D. Shapiro[1], Pam R. Taub[2], Michael J. Louie[3], Lei Lei[3], Christie M. Ballantyne[4]

[1]Center for Prevention of Cardiovascular Disease, Wake Forest University School of Medicine, Winston-Salem, NC; [2]Department of Cardiovascular Medicine, University of California San Diego School of Medicine, La Jolla, CA; [3]Esperion Therapeutics, Inc., Ann Arbor, MI; [4]Department of Medicine, Baylor College of Medicine, Houston, TX

Bempedoic acid was generally well tolerated and led to glycemic parameter improvement and greater LDL-C reduction among patients with metabolic syndrome in this analysis

Background

- Metabolic syndrome (MetS) is characterized by ≥3 of the 5 following criteria[1]:
 - Increased waist circumference
 - Triglycerides (TG) ≥150 mg/dL
 - High-density lipoprotein cholesterol (HDL-C) <40 mg/dL (men) or <50 mg/dL (women)
 - Blood pressure ≥130/85 mmHg
 - Fasting plasma glucose (FPG) ≥100 mg/dL
- MetS has been associated with a 2-fold increased risk of developing atherosclerotic cardiovascular disease (ASCVD)[2]
- Bempedoic acid is an oral ATP-citrate lyase inhibitor that is indicated in the US as an adjunct to diet and maximally tolerated statin therapy, for the treatment of adults with heterozygous familial hypercholesterolemia or established ASCVD who require additional lowering of low-density lipoprotein cholesterol (LDL-C)[3–5]
 - The effect of bempedoic acid as a lipid-lowering therapy (LLT) in patients with MetS has not been established

Objective

- To investigate the efficacy and safety of bempedoic acid, and the effect of bempedoic acid on glycemic parameters, by baseline MetS status

Method

Study design, patients, and treatment

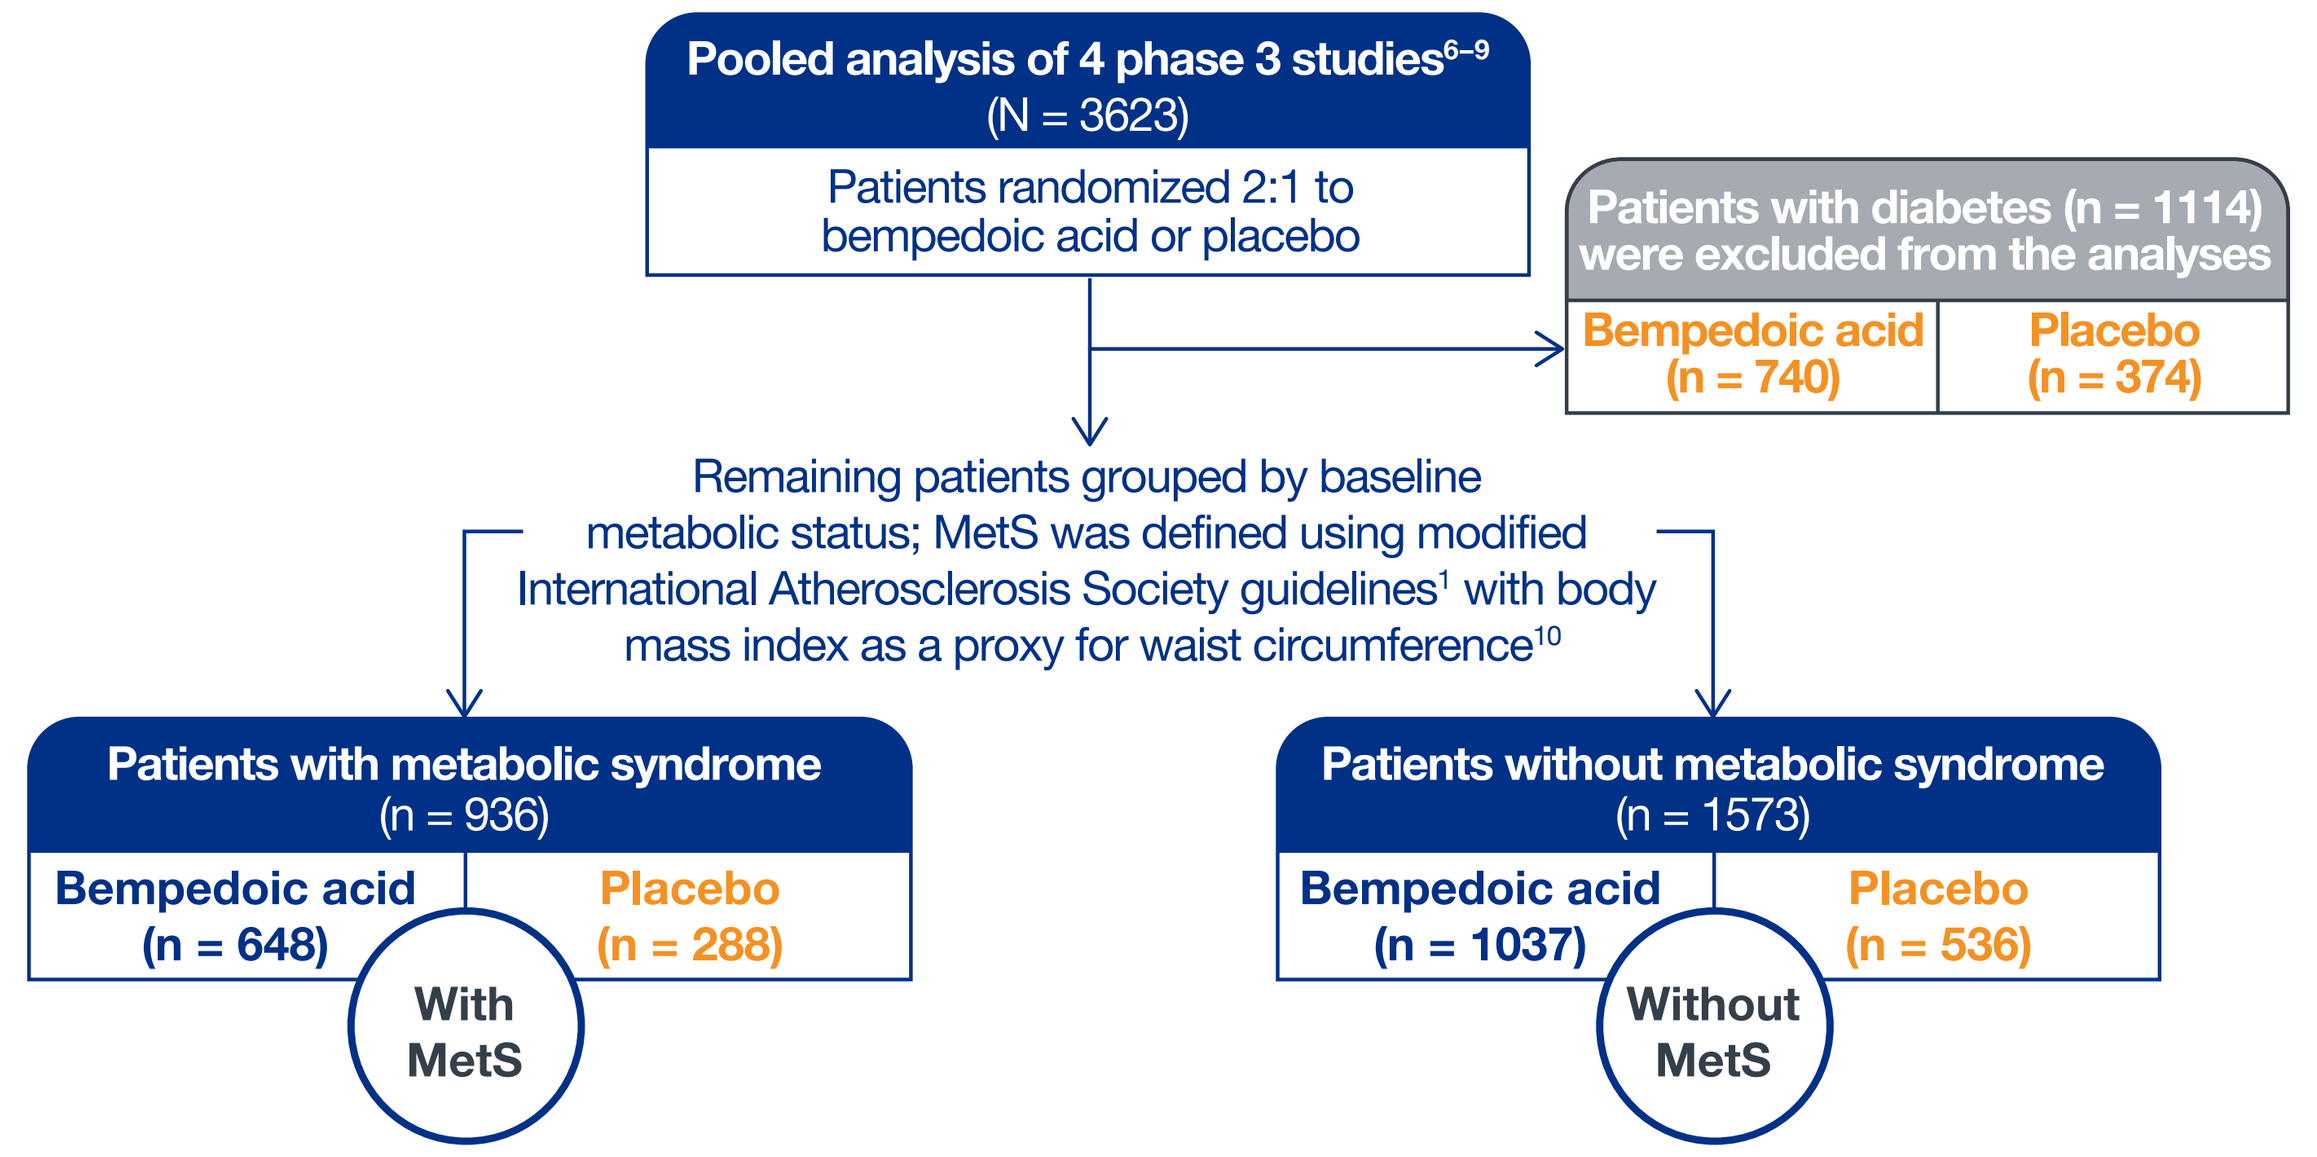
Pooled analysis of 4 phase 3 studies[6–9]
(N = 3623)
Patients randomized 2:1 to bempedoic acid or placebo
Patients with diabetes (n = 1114) were excluded from the analyses
Bempedoic acid (n = 740)
Placebo (n = 374)
Remaining patients grouped by baseline metabolic status; MetS was defined using modified International Atherosclerosis Society guidelines[1] with body mass index as a proxy for waist circumference[10]
Patients with metabolic syndrome (n = 936)
Bempedoic acid (n = 648)
Placebo (n = 288)
With MetS
Patients without metabolic syndrome (n = 1573)
Bempedoic acid (n = 1037)
Placebo (n = 536)
Without MetS

Assessments

- Percent change in LDL-C at week 12
 - LDL-C values were calculated using the Friedewald formula or measured directly using the Multigent Direct LDL assay (Architect system, Abbott) when TG >400 mg/dL or LDL-C <50 mg/dL
- Percent changes in total cholesterol (TC), non–HDL-C, apolipoprotein B (Apo B), high-sensitivity C-reactive protein (hsCRP), TG, and changes in FPG and hemoglobin A1c (HbA1c) at week 12
- Quantification of lipids and biomarkers was performed at a central laboratory (Q2 Solutions)
- Safety assessments were investigator reported and included treatment-emergent adverse events (TEAEs) and serious TEAEs

Statistical analysis

- The efficacy analysis population included all randomized patients; the safety analysis population included all patients receiving at least one dose of study drug
- Least squares (LS) mean difference and its 95% confidence interval (CI), and *P* values within each MetS status were based on an analysis of covariance (ANCOVA)
 - Percent change or change from baseline were the dependent variables, study and treatment were fixed factors, and baseline was a covariate
 - Comparisons of hsCRP were based on a nonparametric method
- The interaction *P* value was obtained from the interaction term via an ANCOVA
 - Percent change or change from baseline (log-transformed for hsCRP) were the dependent variables, study, treatment, MetS (yes/no), and interaction term between treatment and MetS status were fixed factors, and baseline was a covariate

Results

Patients

- Patient characteristics and laboratory parameters at baseline were similar across treatment groups
 - Expected differences between patients with and without MetS were observed (**Table 1**)

Table 1. Demographics and Baseline Characteristics

	With MetS		Without MetS	
Parameter	Bempedoic acid (N = 648)	Placebo (N = 288)	Bempedoic acid (N = 1037)	Placebo (N = 536)
Age, years, mean ± SD	63.7 ± 9.1	64.2 ± 9.0	65.8 ± 9.7	66.2 ± 9.3
Male, % (n)	67.3 (436)	67.7 (195)	65.8 (682)	64.2 (344)
White, % (n)	95.7 (620)	96.2 (277)	96.0 (996)	93.7 (502)
Black, % (n)	2.8 (18)	2.1 (6)	2.9 (30)	4.5 (24)
History of hypertension, % (n)	88.3 (572)	89.2 (257)	64.3 (667)	66.0 (354)
History of ASCVD, % (n)	84.6 (548)	84.7 (244)	84.0 (871)	84.5 (453)
BMI, kg/m², mean ± SD	32.0 ± 4.4	31.9 ± 4.8	27.2 ± 3.9	27.5 ± 3.9
Baseline statin intensity, % (n)				
High	44.0 (285)	42.7 (123)	42.7 (443)	42.7 (229)
Moderate	29.8 (193)	32.3 (93)	32.8 (340)	32.3 (173)
Low	4.6 (30)	4.2 (12)	5.5 (57)	5.8 (31)
None	21.6 (140)	20.8 (60)	19.0 (197)	19.2 (103)
LDL-C, mg/dL, mean ± SD	119.1 ± 38.0	119.3 ± 40.2	114.4 ± 37.3	112.1 ± 36.1
Total cholesterol, mg/dL, mean ± SD	199.8 ± 46.0	200.0 ± 47.8	193.1 ± 43.4	190.7 ± 43.1
Non–HDL-C, mg/dL, mean ± SD	155.0 ± 44.4	155.0 ± 46.2	137.4 ± 40.4	135.1 ± 38.9
HDL-C, mg/dL, mean ± SD	44.7 ± 10.0	45.0 ± 9.9	55.7 ± 13.2	55.5 ± 14.7
Apolipoprotein B, mg/dL, mean ± SD	110.3 ± 33.6	110.8 ± 34.9	97.3 ± 29.0	94.9 ± 28.2
Triglycerides, mg/dL, median (IQR)	170.5 (132.0, 223.0)	172.8 (135.0, 218.2)	110.5 (88.0, 136.0)	108.5 (86.0, 134.0)
hsCRP, mg/L, median (IQR)	2.1 (1.0, 4.1)	2.4 (1.2, 4.7)	1.3 (0.7, 2.7)	1.3 (0.7, 2.5)
HbA1c, %, mean ± SD	5.8 ± 0.3	5.8 ± 0.3	5.6 ± 0.3	5.6 ± 0.3
FPG, mg/dL, mean ± SD	102.8 ± 11.1	103.1 ± 11.2	94.6 ± 9.3	94.5 ± 9.3

ASCVD = atherosclerotic cardiovascular disease; BMI = body mass index; FPG = fasting plasma glucose; HbA1c = hemoglobin A1c; HDL-C = high-density lipoprotein cholesterol; hsCRP = high-sensitivity C-reactive protein; IQR = interquartile range; LDL-C = low-density lipoprotein cholesterol; MetS = metabolic syndrome; SD = standard deviation.

Percent change in LDL-C

- The placebo-corrected LS mean in LDL-C percent change at week 12 was significant for bempedoic acid (*P* <0.0001) regardless of MetS status and was significantly (interaction *P* = 0.0472) more pronounced among patients with MetS compared with patients without MetS (**Figure 1**)

Figure 1. LDL-C Mean Percent Change

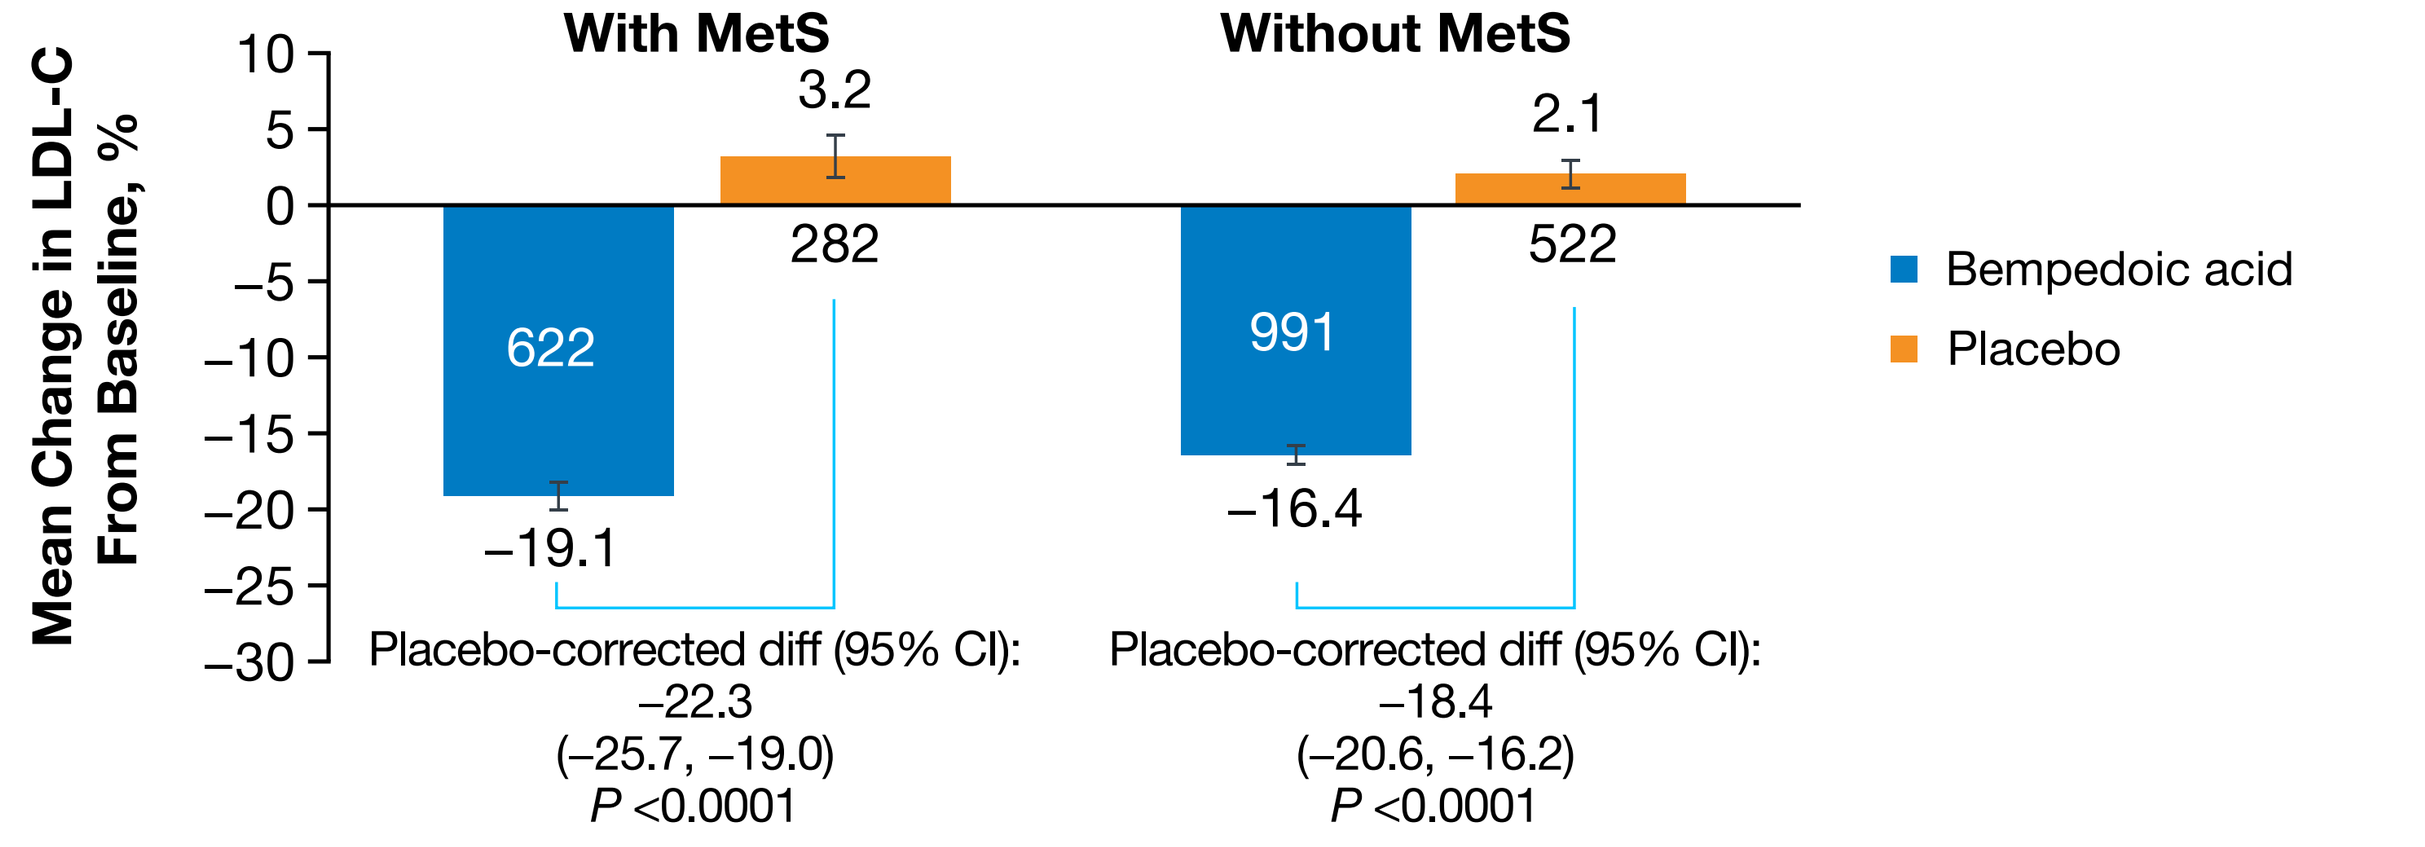
Mean Change in LDL-C From Baseline, %
10
5
0
−5
−10
−15
−20
−25
−30
With MetS
Without MetS
3.2
282
622
−19.1
2.1
522
991
−16.4
Placebo-corrected diff (95% CI): −22.3 (−25.7, −19.0) P <0.0001
Placebo-corrected diff (95% CI): −18.4 (−20.6, −16.2) P <0.0001
Bempedoic acid
Placebo

CI = confidence interval; LDL-C = low-density lipoprotein cholesterol; MetS = metabolic syndrome; SD = standard deviation.

Change in TC, non–HDL-C, TG, Apo B, hsCRP, HbA1c, and FPG

- The placebo-corrected LS mean in percent change at week 12 for TC, non–HDL-C, TG, and Apo B were significant (*P* ≤0.02) for patients with and without MetS (interaction *P* = 0.523, *P* = 0.451, *P* <0.0001, and *P* = 0.530, respectively) (**Table 2**)
- The median difference at week 12 for hsCRP was significant for patients with and without MetS (with MetS, –17.9 [–25.1, –10.7], *P* <0.0001; without MetS, –19.9 [–26.5, –13.2], *P* <0.0001; interaction *P* = 0.965 in the log scale) (**Table 2**)
- The placebo-corrected LS mean percent change at week 12 for HbA1c and FPG was significant for patients with MetS (HbA1c, –0.07 [–0.10, –0.04], *P* <0.0001; FPG, –2.4 [–4.0, –0.9], *P* = 0.002) and insignificant for patients without MetS (interaction *P* = 0.0003 and *P* = 0.002, respectively) (**Table 2**)

Table 2. Percent Change from Baseline to Week 12 for lipid and glycemic parameters

		With MetS		Without MetS	
Parameter		Bempedoic acid (N = 648)	Placebo (N = 288)	Bempedoic acid (N = 1037)	Placebo (N = 536)
TC	(n)	624	282	992	522
	LS mean (SE)	−11.7 (0.7)	1.5 (0.9)	−11.1 (0.4)	1.3 (0.6)
	Placebo-corrected LS mean (95% CI)	−13.2 (−15.5, −11.0)		−12.4 (−13.9, −10.9)	
	P value	*P* <0.0001		*P* <0.0001	
Non–HDL-C	(n)	623	282	991	522
	LS mean (SE)	−12.8 (0.8)	1.9 (1.2)	−13.3 (0.5)	2.6 (0.9)
	Placebo-corrected LS mean (95% CI)	−14.7 (−17.6, −11.8)		−15.9 (−18.0, −13.9)	
	P value	*P* <0.0001		*P* <0.0001	
TG	(n)	624	282	992	522
	LS mean (SE)	13.9 (2.1)	0.7 (1.9)	4.5 (1.2)	8.9 (1.5)
	Placebo-corrected LS mean (95% CI)	13.1 (7.5, −18.7)		−4.4 (−8.1, − 0.7)	
	P value	*P* <0.0001		*P* = 0.02	
Apo B	(n)	610	276	977	514
	LS mean (SE)	−10.7 (0.7)	2.8 (1.1)	−10.5 (0.5)	4.0 (0.8)
	Placebo-corrected LS mean (95% CI)	−13.6 (−16.2, −10.9)		−14.5 (−16.4, −12.6)	
	P value	*P* <0.0001		*P* <0.0001	
hsCRP	(n)	605	275	976	515
	Median (IQR)	−26.9 (59.3)	−5.9 (77.5)	−15.8 (80.0)	4.8 (87.9)
	Median difference (95% CI)	−17.9 (−25.1, −10.7)		−19.9 (−26.5, −13.2)	
	P value	*P* <0.0001		*P* <0.0001	
HbA1c	(n)	615	281	987	522
	LS mean (SE)	−0.06 (0.01)	0.01 (0.01)	−0.02 (0.01)	−0.02 (0.01)
	Placebo-corrected LS mean (95% CI)	−0.07 (−0.10, −0.04)		0.00 (−0.02, 0.02)	
	P value	*P* <0.0001		*P* = 0.95	
FPG	(n)	622	282	990	521
	LS mean (SE)	−0.4 (0.4)	2.0 (0.7)	1.2 (0.2)	1.0 (0.4)
	Placebo-corrected LS mean (95% CI)	−2.4 (−4.0, −0.9)		0.2 (−0.7, 1.0)	
	P value	*P* = 0.002		*P* = 0.68	

Apo B = apolipoprotein B; CI = confidence interval; FPG = fasting plasma glucose; HbA1c = hemoglobin A1c; HDL-C = high-density lipoprotein cholesterol; hsCRP = high-sensitivity C-reactive protein; IQR = interquartile range; LS = least squares; MetS = metabolic syndrome; SE = standard error; TC = total cholesterol; TG = triglycerides.

Safety

- The bempedoic acid safety profile was similar in patients with or without MetS, and the incidence of TEAEs was similar between the placebo and BA groups (**Table 3**)

Table 3. Treatment-Emergent Adverse Events

	With MetS		Without MetS	
Parameter, % (n)	Bempedoic acid (N = 648)[a]	Placebo (N = 288)[a]	Bempedoic acid (N = 1036)[a]	Placebo (N = 535)[a]
Any TEAE	70.8 (459)	67.4 (194)	74.7 (774)	72.7 (389)
Serious TEAE	12.5 (81)	12.5 (36)	14.1 (146)	11.6 (62)
TEAE related to study drug	25.0 (162)	18.4 (53)	24.6 (255)	20.6 (110)
TEAE leading to discontinuation	12.2 (79)	6.6 (19)	11.2 (116)	7.3 (39)
TEAE with fatal outcome	0.2 (1)	0.3 (1)	0.9 (9)	0.2 (1)
Most common TEAEs,[b]				
Nasopharyngitis	5.9 (38)	9.7 (28)	8.8 (91)	10.3 (55)
Myalgia	4.5 (29)	3.5 (10)	5.7 (59)	6.0 (32)
Arthralgia	3.9 (25)	5.2 (15)	4.7 (49)	4.7 (25)
Blood CPK increased	3.7 (24)	2.1 (6)	1.4 (14)	0.9 (5)
Headache	3.1 (20)	3.1 (9)	2.5 (26)	3.4 (18)
Muscle spasms	3.1 (20)	3.1 (9)	4.5 (47)	2.1 (11)
Pain in extremity	3.1 (20)	1.4 (4)	3.4 (35)	2.2 (12)
Upper respiratory tract infection	3.1 (20)	3.1 (9)	4.2 (44)	4.7 (25)
Urinary tract infection	2.8 (18)	3.1 (9)	4.9 (51)	6.0 (32)
Diarrhea	2.8 (18)	2.8 (8)	3.7 (38)	2.1 (11)
Dizziness	2.6 (17)	1.7 (5)	3.7 (38)	3.4 (18)
Back pain	1.9 (12)	2.4 (7)	3.4 (35)	2.8 (15)

[a]N numbers correspond to the safety-evaluable population. [b]Most common TEAEs were defined as those occurring in ≥3% of patients in any bempedoic acid treatment group. CPK = creatine phosphokinase; MetS = metabolic syndrome; TEAE = treatment-emergent adverse event.

Conclusions

- **Bempedoic acid was associated with significant decreases in LDL-C, TC, non–HDL-C, Apo B, and hsCRP regardless of the presence or absence of MetS**
 - **Bempedoic acid lowered LDL-C levels to a greater extent in patients with MetS compared with patients without MetS**
- **Slight reductions in both HbA1c and FPG were noted with bempedoic acid among patients with MetS**
 - **High hsCRP levels have been associated with impaired insulin sensitivity[11]**
 - **Patients with MetS had higher baseline hsCRP and a similar percent decrease in hsCRP with bempedoic acid compared with patients without MetS**
- **Bempedoic acid was generally well tolerated with comparable safety profiles in patients with and without MetS**

References

1. Expert Dyslipidemia Panel of the International Atherosclerosis Society. *J Clin Lipidol*. 2014;8:29–60.
2. Alberti KGMM, et al. *Circulation*. 2009;120:1640–1645.
3. Banach M, et al. *JAMA Cardiol*. 2020;5:1124–1135.
4. Bays HE, et al. *J Clin Lipidol*. 2020;14:649–659.
5. NEXLETOL® (bempedoic acid) [package insert]. Ann Arbor, MI: Esperion Therapeutics, Inc.; September 2021.
6. Ray KK, et al. *N Engl J Med*. 2019;380:1022–1032.
7. Laufs U, et al. *J Am Heart Assoc*. 2019;8:e011662.
8. Ballantyne CM, et al. *Atherosclerosis*. 2018;277:195–203.
9. Goldberg AC, et al. *JAMA*. 2019;322:1780–1788.
10. Henry RR, et al. *Diabetes Obes Metab*. 2018;20:1632–1641.
11. Ndumele CE, et al. *J CardioMetab Syndr*. 2006;1:190–196.

Acknowledgments and Disclosures

We thank all investigators and patients who participated in these trials. Clinical trials included in this analysis were funded by Esperion Therapeutics, Inc. Esperion participated in the study design, research, data collection, analysis and interpretation of data, and writing, reviewing, and approving the poster for presentation. All authors had access to the data and participated in the development, review, and approval of the poster. Medical writing and editing assistance were provided by Katerina Pipili, PhD, and Charlotte Watson, of Spark Medica Inc., funded by Esperion Therapeutics, Inc.

MDS has received funding/grant support from Amgen, the National Institutes of Health, and Novartis, and honoraria for consultancy from Amgen, Novartis, Novo Nordisk, and Regeneron. PT has received funding/grant support from the American Heart Association, the National Institutes of Health, and the Federal Emergency Management Agency, and honoraria for consultancy from Amgen, Bayer, Boehringer Ingelheim, Esperion, Medtronic, and Novo Nordisk. MJL is a current employee of Esperion Therapeutics, Inc., and may own Esperion stock and/or stock options. LL is a consultant for Esperion Therapeutics, Inc. CMB has received research grant(s)/support paid to his institution from Abbott Diagnostics, Akcea, Amarin, Amgen, Esperion, Ionis, Novartis, Regeneron, Roche Diagnostics, Sanofi Synthelabo, the National Institutes of Health, the American Heart Association, and the American Diabetes Association; he has also served as a consultant for Abbott Diagnostics, Amarin, Amgen, AstraZeneca, Boehringer Ingelheim, Eli Lilly, Esperion, Intercept, Ionis, Matinas BioPharma, Inc., Merck, Novartis, Novo Nordisk, Regeneron, Roche Diagnostics, and Sanofi Synthelabo.

Presented at the Cardiometabolic Health Congress, October 19–22, 2022.